Design of a Cross-Sectional Observational Study of Caregiver Burden of Patients who are Diagnosed with Multiple Sclerosis or are Post-Stroke with Upper and/or Lower-Limb Spasticity in the US

Elson CL Pharm.D. Candidate[1]; Ertl J M.A.[2] ; Burk CT Pharm.D., M.S.[3]; Varon SF Ph.D.[3]

[1]University of Georgia, College of Pharmacy, Athens, GA; [2]KantarHealth, Princeton, NJ; [3]Allergan Inc., Irvine, CA

BACKGROUND

- Spasticity can occur following central nervous system (CNS) injury, such as in stroke and multiple sclerosis (MS).
- Spasticity is believed to occur in approximately 18% (Lundstrom et al) of post-stroke survivors and 67% (Rizzo et al) of multiple sclerosis patients; disabling spasticity occurs in 4% of both groups. Very often, care giving of these patients becomes the responsibility of a family member or close relative (Khan et al).
- Tasks associated with caring for a patient with spasticity due to MS or stroke include feeding, dressing, bathing and toileting (Pakenham et al), among other activities. Completing these activities- especially if the caregiver's health is impaired or the caregiver is currently working full-time can negatively affect the health-related quality of life (HRQoL) and work productivity of the caregiver, indicating an increased caregiver burden.
- In previous research, caregivers of patients with these conditions have reported depression, negative consequences on social life and coping ability, resulting in social, financial and emotional burden (Puymbroeck and Rittman).
- Limited research has been conducted to quantify the magnitude and impact that caring for a spasticity patient exerts on the caregiver warranting further research to better understand the effects on HRQoL in caregivers of patients in these populations.

STUDY OBJECTIVES

The objective of this study is to determine the effect upper-limb (UL) and/or lower-limb (LL) spasticity due to stroke or MS has on:

1) Caregiver's HRQoL and emotional status;
2) Caregiver time, time lost from work;
3) Patient's utilization of healthcare resources from a caregiver's perspective;
4) Caregiver's perception of patient satisfaction and adverse effects associated with current spasticity medications.

STUDY DESIGN

- Web-based, cross-sectional observational study conducted in the United States.
- Caregiver respondents for this study will be recruited from members of the Lightspeed Research (LSR) panel who have previously taken the National Health and Wellness Survey (NHWS) and responded that they were caregivers for post stroke or MS patients, and from the LSR panel at large.
- Respondents will be provided consent via "opting-in" using a web link as a proxy for written consent and will be directed to the online study.
- Respondents will be required to complete a screener questionnaire to determine eligibility. Respondents who qualify to participate in this study will be requested to complete the Disability Assessment Scale (DAS), Short Form 12 (SF-12), Patient Health Questionnaire (PHQ-4), Zarit Burden Interview (BI), Oberst Caregiving Burden Scale (OCBS), Work Productivity and Activity Index – Informal Caregiving (WPAI-CG), and Caregiver Strain Index (CSI).
- Target sample is approximately 300 caregivers of patients with UL and / or LL spasticity:
 - 150 caregivers of post-stroke
 - 150 caregivers of MS patients
- Prior to e-mailing invitation to all eligible members from the previously identified NHWS and LSR panel, the survey will be fielded to approximately 10% of the planned field sample to determine if any logistical problems exist and to confirm estimated response rate. If logistical problems are found these will be corrected or if response rate is inadequate then multiple reminders will be sent or an increase in responders' incentives will be considered prior to survey launch to the remaining 90% of targeted panelists.

STUDY POPULATION

- The US National Health and Wellness Survey (NHWS) is an annual study of the healthcare attitudes and behaviors of nationally representative samples of the adult populations of the United States. In 2008, the US NHWS was fielded to 63,000 members of the general panel of LSR; in 2009, the survey has been completed by 75,000 respondents. Through panel identification numbers respondents can be identified and recontacted for additional studies such as this one.
- Approximately 1288 post-stroke and 353 MS caregiver respondents of the NHWS in either 2007, 2008 or 2009 and panelists from the LSR panel at large will be invited to participate in the study. It is anticipated that approximately 166 of the 300 total respondents from panelists who participated in the NHWS in the years 2007-2009 will be recruited. The remaining respondents will be supplemented from the LSR panel at large.

Inclusion Criteria
- Adult 18 years or older
- Currently caring for a patient who has been identified as: suffering stroke or diagnosed with MS more than 3 months ago
- Has not participated in a similar study in the past 3 months
- Resides in the United States
- Reads and writes English
- Provides consent to participate in the study

Exclusion Criteria
- A formal or paid caregiver

FIGURE 1

Diagram of Estimated Survey Target Sample

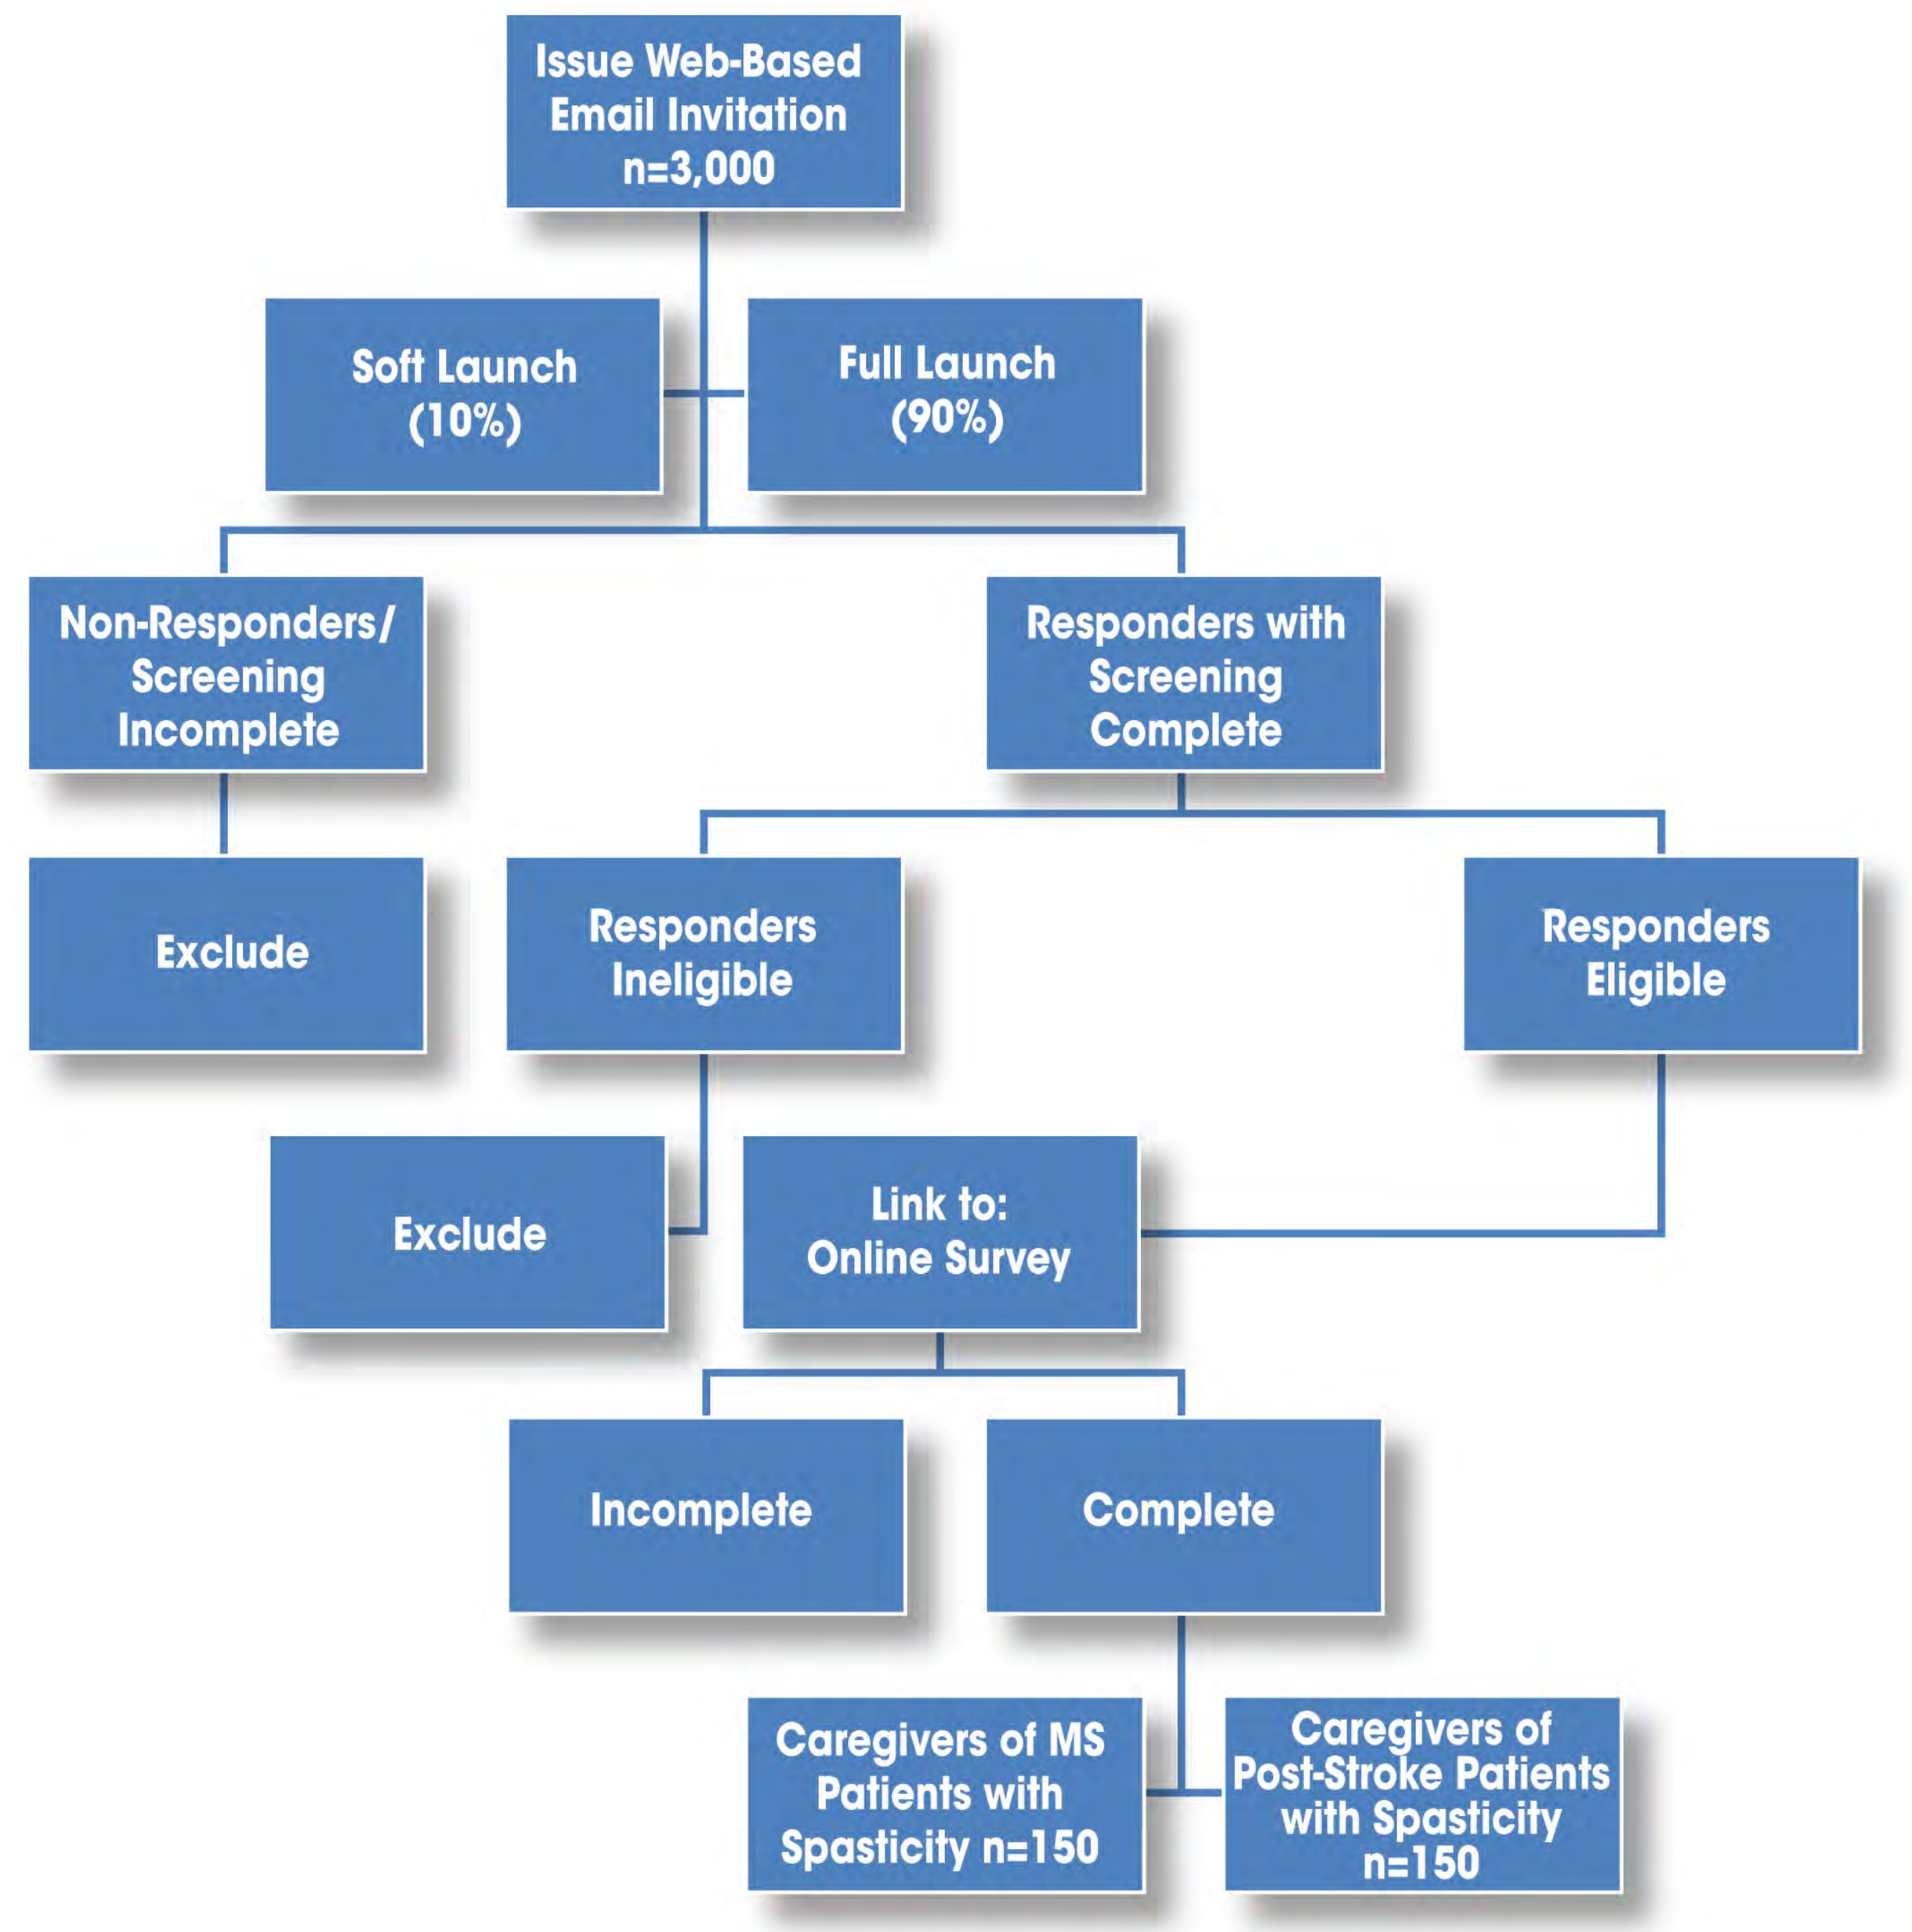
Issue Web-Based Email Invitation n=3,000
Soft Launch (10%)
Full Launch (90%)
Non-Responders/ Screening Incomplete
Responders with Screening Complete
Exclude
Responders Ineligible
Responders Eligible
Exclude
Link to: Online Survey
Incomplete
Complete
Caregivers of MS Patients with Spasticity n=150
Caregivers of Post-Stroke Patients with Spasticity n=150

DATA COLLECTION

The following data will be collected: caregiver and patient demographic information, conditions/comorbidities of caregiver and patient, caregiver's tasks and responsibilities, effects of caregiving on the caregiver's health and other activities, clinical characteristics of patient, and resource utilization of patient.

TABLE 1

Survey instruments utilized in the study to capture endpoints of study

Indirect Patinet Information	Direct Caregiver Information
Disability Assessment Scale (DAS)	Short Form-12 (SF-12)
Individual Questions	Patient Health Questionnaire (PHQ-4)
• Condition of caregiver's patient • Characteristics, history and severity of patient's spasticity • Patient's resource use and satisfaction with spasticity treatment	Caregiver Burden Instruments
	• Oberst Caregiving Burden Scale (OCBS) • Zarit Burden Interview (BI) • Caregiver Strain Index (CSI)
	Work Productivity and Activity Index - Informal Caregiving (WPAI-CG)
	Individual Questions
	• Demographic information of caregiver • Conditions/comorbidities of caregiver • Charcteristics of caregiving • Effects of caregiving on health and other activities • Resources available to caregiver

RESULTS

Caregiver recruitment will begin in October 2009 and it is projected to conclude in November 2009 after running for three weeks. Data processing will occur for approximately 1 week. Analysis and publication will then follow.

CONCLUSIONS

This study will provide insight into the burden experienced by caregivers of patients with UL and/or LL spasticity and will help prepare caregivers for the additional responsibilities and strain they may experience as a result of their responsibilities as caregivers. Additionally, this study will provide a better understanding of resource utilization and the satisfaction with spasticity therapy, which will aid in spasticity therapy management.

REFERENCES

1. Khan F, Pallant J, Brand C. Caregiver strain and factors associated with caregiver self-efficacy and quality of life in a community cohort with multiple sclerosis. Disability and Rehabilitation, August 2007; 29(16): 1241 – 1250.
2. Lundstrom E, Terent A, Borg J. Prevalence of disabling spasticity 1 year after first-ever stroke. European Journal of Neurology 2008, 15: 533–539.
3. Pekenham, KI. The nature of caregiving in multiple sclerosis: development of the caregiving tasks in multiple sclerosis scale. Multiple Sclerosis 2007; 13: 929–938.
4. Rizzo MS, Hadjimichael OC, Preiningerova J, Vollmer TL. Prevalence and treatment of spasticity reported by multiple sclerosis patients. Multiple Sclerosis 2004; 10: 589-595.
5. Van Puymbroeck M, Rittman M. Quality of life predictors for caregivers at 1 and 6 months poststroke: results of path analyses. J Rehabil Res Dev 2005; 42:747-60.

ACKNOWLEDGEMENTS

This work was fully supported by Allergan Inc., Irvine, California. We acknowledge the assistance of KantarHealth, Priceton, NJ, in the design and implementation of this study. We would like to specifically acknowlege the following KantarHealth members involved in this project including:

- David A. Pomerantz, SVP, Business Development, who is responsible for the business development team in the Health Sciences Practice at KantarHealth.
- Ariella S. Dugan, Director, Research Services, serving as the operations project oversight lead.
- Marco DiBonaventura, Ph.D., Manager, Health Economics and Outcomes Research, serving as the leader of analytic efforts and advanced statistical analysis.